Safety and Feasibility of Intranigral and Intraputaminal CERE-120 (AAV2-neurturin) administration in Parkinson's Disease

J. Siffert[1], M. A. Stacy[2], D. Turner[2], M. Tagliati[3], R. Alterman[3], S.A. Factor[4], N. Boulis[4] and C. W. Olanow[3, 6], A.E. Lang[5], A.M. Lozano[5] and R.T. Bartus[1]

[1]Ceregene, Inc., San Diego CA, US A; [2]Duke University, Durham, NC, USA; [3]Mount Sinai School of Medicine, New York, NY, USA; [4]Emory University School of Medicine, Atlanta, GA, USA; [5]University of Toronto, Toronto, Canada; and [6]Institute of Neurology, IRCCS San Raffaele Pisana, Rome, Italy

Abstract

Objective:
To evaluate the feasibility and safety of CERE-120 administration to substantia nigra (SN) and putamen in Parkinson's disease (PD) in an open-label Phase-1trial.
Background:
CERE-120 employs gene transfer to deliver neurturin (NRTN), a neurotrophic-factor that enhances dopaminergic neuron function and survival in animal models of PD. Intraputaminal CERE-120 in 50 PD patients followed postoperatively for 3-5+ years appears safe. Whereas the completed double-blind, sham-surgery-controlled Phase-2a study failed to meet the primary-endpoint (UPDRS motor "off") at 12-months, the primary endpoint was reached for a subset of patients with double-blind assessments up to 18-months, as were several secondary endpoints, with no endpoint favoring sham-surgery (Marks et al, 2010, Lancet Neurol). Analysis of PD brain tissue post-CERE-120 in 3 patients showed that although NRTN is expressed in the putamen, minimal NRTN was transported to SN pars compacta, where it is required for induction of repair genes (e.g., Bartus et al, 2010, Mov Disorders). Following successful completion of nonclinical safety/feasibility tests, we are now testing CERE-120 delivered directly to the SN, combined with a higher dose to the putamen, to maximize the bioactivity.
Methods
Phase-1 open-label, two dose-cohorts (N=3, each). Both cohorts received $2.0\text{x}10^{11}$vector genomes (vg) into each SN; dose-cohort-1 also received into each putamen $2.7\text{x}10^{11}$vg (same as Phase-2a study) while dose-cohort-2 received $10\text{x}10^{11}$vg. Patients had idiopathic PD with robust response to levodopa but with motor-complications. The primary-endpoint was safety/tolerability.
Results
Each patients was dosed a month apart between 12/2009-6/2010. No surgical complications, serious adverse events or weight-loss occurred. Brain MRI at postoperative days 1 and 30 revealed no unexpected changes. Compared to baseline, UPDRS motor scores off medication improved an average of 22% at 9 months (N=6) and 19% at 12 months (N=3) (*updated as of March, 2011*)
Interpretation
Preliminary data supports safety and feasibility of intranigral and intraputaminal CERE-120 administration in this small study. A sham-surgery-controlled Phase-2b study is ongoing.

Background and Objectives

- AAV2-NRTN (CERE-120), an AAV2 vector that delivers human NRTN, is being developed as a potential therapy for Parkinson's disease (PD) by Ceregene, Inc.
- NRTN, like GDNF, can protect nigrostriatal dopamine neurons from degeneration, restore function and thus has potential as a novel therapeutic for PD patients.
- Intraputaminal administration of CERE-120 in a Phase 1 (n=12) and a Phase 2a study (n=38) in advanced PD patients appears safe with follow up of 60+ months (Marks et al., 2008 and 2010).
- Modest benefits were seen in the completed Phase 2a study where 58 subjects were randomized to either receive CERE-120 or undergo a sham surgery procedure (**Figure 1**)
- Recent brain autopsy of PD patients treated with intraputaminal AAV2-NRTN (Bartus et al.) indicate that only limited amounts of NRTN are transported from the targeted striatum to the cell bodies in the SN, likely due to impaired axonal transport in PD.
- Before pursuing dual intranigral and intraputaminal dosing in the clinic, a series of nonclinical experiments were conducted to test the safety of NRTN expression in and around the SN (**Tables 1 and 2**) as well as potential efficacy (**Figure 2**) using a 200-fold dose range of CERE-120.

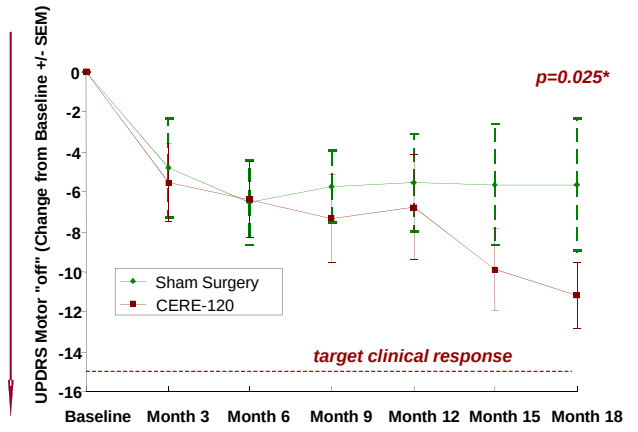
UPDRS Motor "off" (Change from Baseline +/- SEM)
0
-2
-4
-6
-8
-10
-12
-14
-16
p=0.025*
Sham Surgery
CERE-120
target clinical response
Baseline
Month 3
Month 6
Month 9
Month 12
Month 15
Month 18

ANCOVA model with a main effect for treatment group and baseline UPDRS Part III motor score in the practically defined off condition as covariate. Note: at 18 mos, 14 subjects have scores; therefore 16 subjects: LOCF

Figure 1. UPDRS Motor "off" score; change from baseline under double blind conditions

Group	# Rats	Injection Vol (µL)	Dose/Hem (vector genomes)	Designated Dose	Dose Description
Formulation Buffer	8	2 or 5	NA	FB	Control
AAV2-NRTN	8	2	$0.32\text{x}10^9$	Dose A	1/5th dose projected to target SN
AAV2-NRTN	8	2	$1.6\text{x}10^9$	Dose B	Dose successfully projected to cover majority of SN
AAV2-NRTN	8	2	$8\text{x}10^9$	Dose C	5X higher dose than required to cover nigra
AAV2-NRTN	8	5	$60\text{x}10^9$	Dose D	~40X higher dose than required to cover nigra
AAV2-GDNF	8	2	$8\text{x}10^9$	GDNF	GDNF dose reported in literature to produce body weight changes

Table 1: Dose response study targeting the SN with a range of AAV2-NRTN doses or formulation buffer (FB) in rats to test the safety and feasibility, as well as spread of protein

MEASUREMENTS	OUTCOMES
In-life Safety and Tolerability	
• Daily cage-side observations • Weekly body weight • Weekly food intake	• No observable differences • Modest reduction in wt gained; highest doses only • Modest reduction in food intake
Neurobehavioral Examination	
• Hand-held examination • Cranial/spinal reflexes • Body temperature • Open field behavior	• No apparent changes • All reflexes intact • No change or difference • No observable difference
Transgene Expression	
• NRTN labeling throughout brain • Quantification of NRTN distribution	• Dose-related spread of NRTN expression from targeted SN • Dose-related increase in volume of NRTN expression
Neurotoxicity	
• H&E staining throughout brain • Blinded histopath, analyses (e.g. neurotoxicity, inflammation, gliosis, cellular proliferation, hemorrhage)	• No evidence of inflammation, histological abnormalities or any other pathological change

Table 2. Endpoints and safety results from dose-response study

Figure 2. Nigral vs. striatal targeting of AAV-NRTN (CERE-120) to degenerating nigrostriatal dopamine neurons: Animals received AAV2-NRTN or FB control into either SN or striatum, simultaneously with an intrastriatal injection of 6-OHDA. They were sacrificed 4 weeks later. Quantitation of TH+ cells in the SN revealed that nigral delivery of AAV2-NRTN resulted in far greater cell sparing (~90% of intact side), compared to striatal delivery (~44% of intact side) in a 6-OHDA paradigm of ongoing degeneration

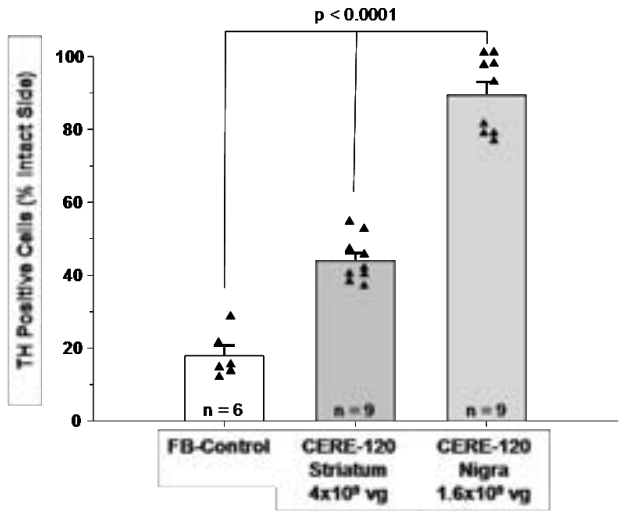
p < 0.0001
TH Positive Cells (% Intact Side)
100
80
60
40
20
0
n = 6
n = 9
n = 9
FB-Control
CERE-120 Striatum 4x10⁹ vg
CERE-120 Nigra 1.6x10⁹ vg

Methods

- Phase-1 open-label, two dose-cohorts (N=3, each)
- Both cohorts received CERE-120 $2.0\text{x}10^{11}$vg into each SN
 - Dose-cohort-1 also received into each putamen $2.7\text{x}10^{11}$vg (same as Phase-2a study)
 - Dose-cohort-2 received $10\text{x}10^{11}$vg
- Eligibility
 - Between 35-70 y with idiopathic PD
 - Robust motor response to levodopa
 - Presence of motor complications (e.g. dyskinesia, wearing off, etc.)
 - No evidence of dementia or "extranigral" symptoms
- Primary-endpoint was safety/tolerability over 1 year

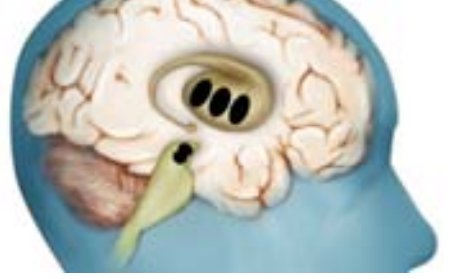

Figure 3. Expected NRTN expression following intraputaminal and intranigral stereotactic CERE-120 delivery. Putamen: 3 needle passes with one drug deposit each and SN: 1 needle pass and two drug deposits

Results

- Patients were dosed monthly between 12/2009-6/2010.
- Follow-up ranges from 9-12 months post-surgery
- No serious adverse events (SAEs) have been reported
- There were no surgical complications or prolonged hospitalizations
- All adverse events were mild/moderate and expected
 - Most were transient
- No weight loss
- Brain MRI at postoperative days 1 and 30, and at 1 year (N=3) revealed no complications
- Compared to baseline, UPDRS motor scores off medication improved an average of 22% at 9 months (N=6) and 19% at 12 months (N=3)

Conclusions

- Preliminary data supports safety and feasibility of intranigral and intraputaminal CERE-120 administration in this small Phase 1 study
- Combined with nonclinical data showing safety at high dose multiples and an incremental benefit from dual administration (nigral + putamen) of CERE-120, Phase 1 data provide strong support for continued CERE-120 clinical development
- A randomized, sham-surgery-controlled, multicenter Phase-2b study in PD (N=52) is now underway at 11 centers across the US

References

1. Marks et al., Lancet Neurology, 2008; vol. 7: 400-08
2. Marks et al., Lancet Neurology, 2010; vol. 9: 1164-72
3. Bartus et al., Movement Disorders, 2011 vol. 26, No.1: 27-36

Partial funding for this study was provided by The Michael J. Fox Foundation for Parkinson's Research (MJFF) as part of the MJFF 2010 LEAPS (Linked Efforts to Accelerate Parkinson's Solutions) program on neurotrophic factors.

Ceregene Inc. 9381 Judicial Drive, San Diego, CA 92121, USA www.ceregene.com